EFFECT OF ALLERGY SPECIALTY CARE ON HEALTHCARE UTILIZATION AMONG CHILDREN WITH PEANUT ALLERGY IN THE UNITED STATES

#120

Matthew Greenhawt,[1] Elissa Abrams,[2] Joseph M. Chalil,[3] Todd D. Green,[3,4] Marcus Shaker[5,6]

[1]Children's Hospital Colorado, Section of Allergy and Immunology, University of Colorado School of Medicine, Aurora, CO, USA; [2]University of Manitoba, Winnipeg, Manitoba, Canada; [3]DBV Technologies, Montrouge, France; [4]UPMC Children's Hospital of Pittsburgh, Pittsburgh, PA, USA; [5]Section of Allergy and Immunology, Dartmouth-Hitchcock Medical Center, Lebanon, NH, USA; [6]Dartmouth Geisel School of Medicine, Hanover, NH, USA

RATIONALE

- Peanut allergy (PA) can lead to severe, potentially life-threatening reactions resulting from unintended exposure to peanut, many of which may require the use of emergency and advanced care services[1,2]
- PA is associated with increased healthcare costs and resource utilization, including a greater number of contacts with primary care providers, inpatient care, prescriptions, outpatient care, and emergency medical services compared with individuals without PA[1]
- The specific influence of allergist care on PA-related healthcare utilization and costs are unknown, as are the differences in costs between PA and other food allergies
- This analysis will aim to expand upon previous findings[1] regarding the overall burden of illness, including treatment patterns, healthcare costs, and resource utilization, among patients with PA versus other food allergies, as well as investigate the impact of allergy specialty care on such resource utilization

OBJECTIVES

- The objectives of this analysis were to assess all-cause and PA-related healthcare resource utilization and costs, as well as incidence of anaphylactic reactions, during a 12-month follow-up period among:
 - o Patients with PA versus patients with non-PA food allergy controls (NPAFAC)
 - o Patients with PA and at least 1 allergist visit versus those with PA but without an allergist visit

METHODS

Design and Patients

- This retrospective, observational study used administrative healthcare claims from the IBM MarketScan® Commercial Claims and Encounters Database (captured from January 1, 2010 through June 30, 2019) (**Figure 1**)

Figure 1. Study Period for Burden of Illness Analysis

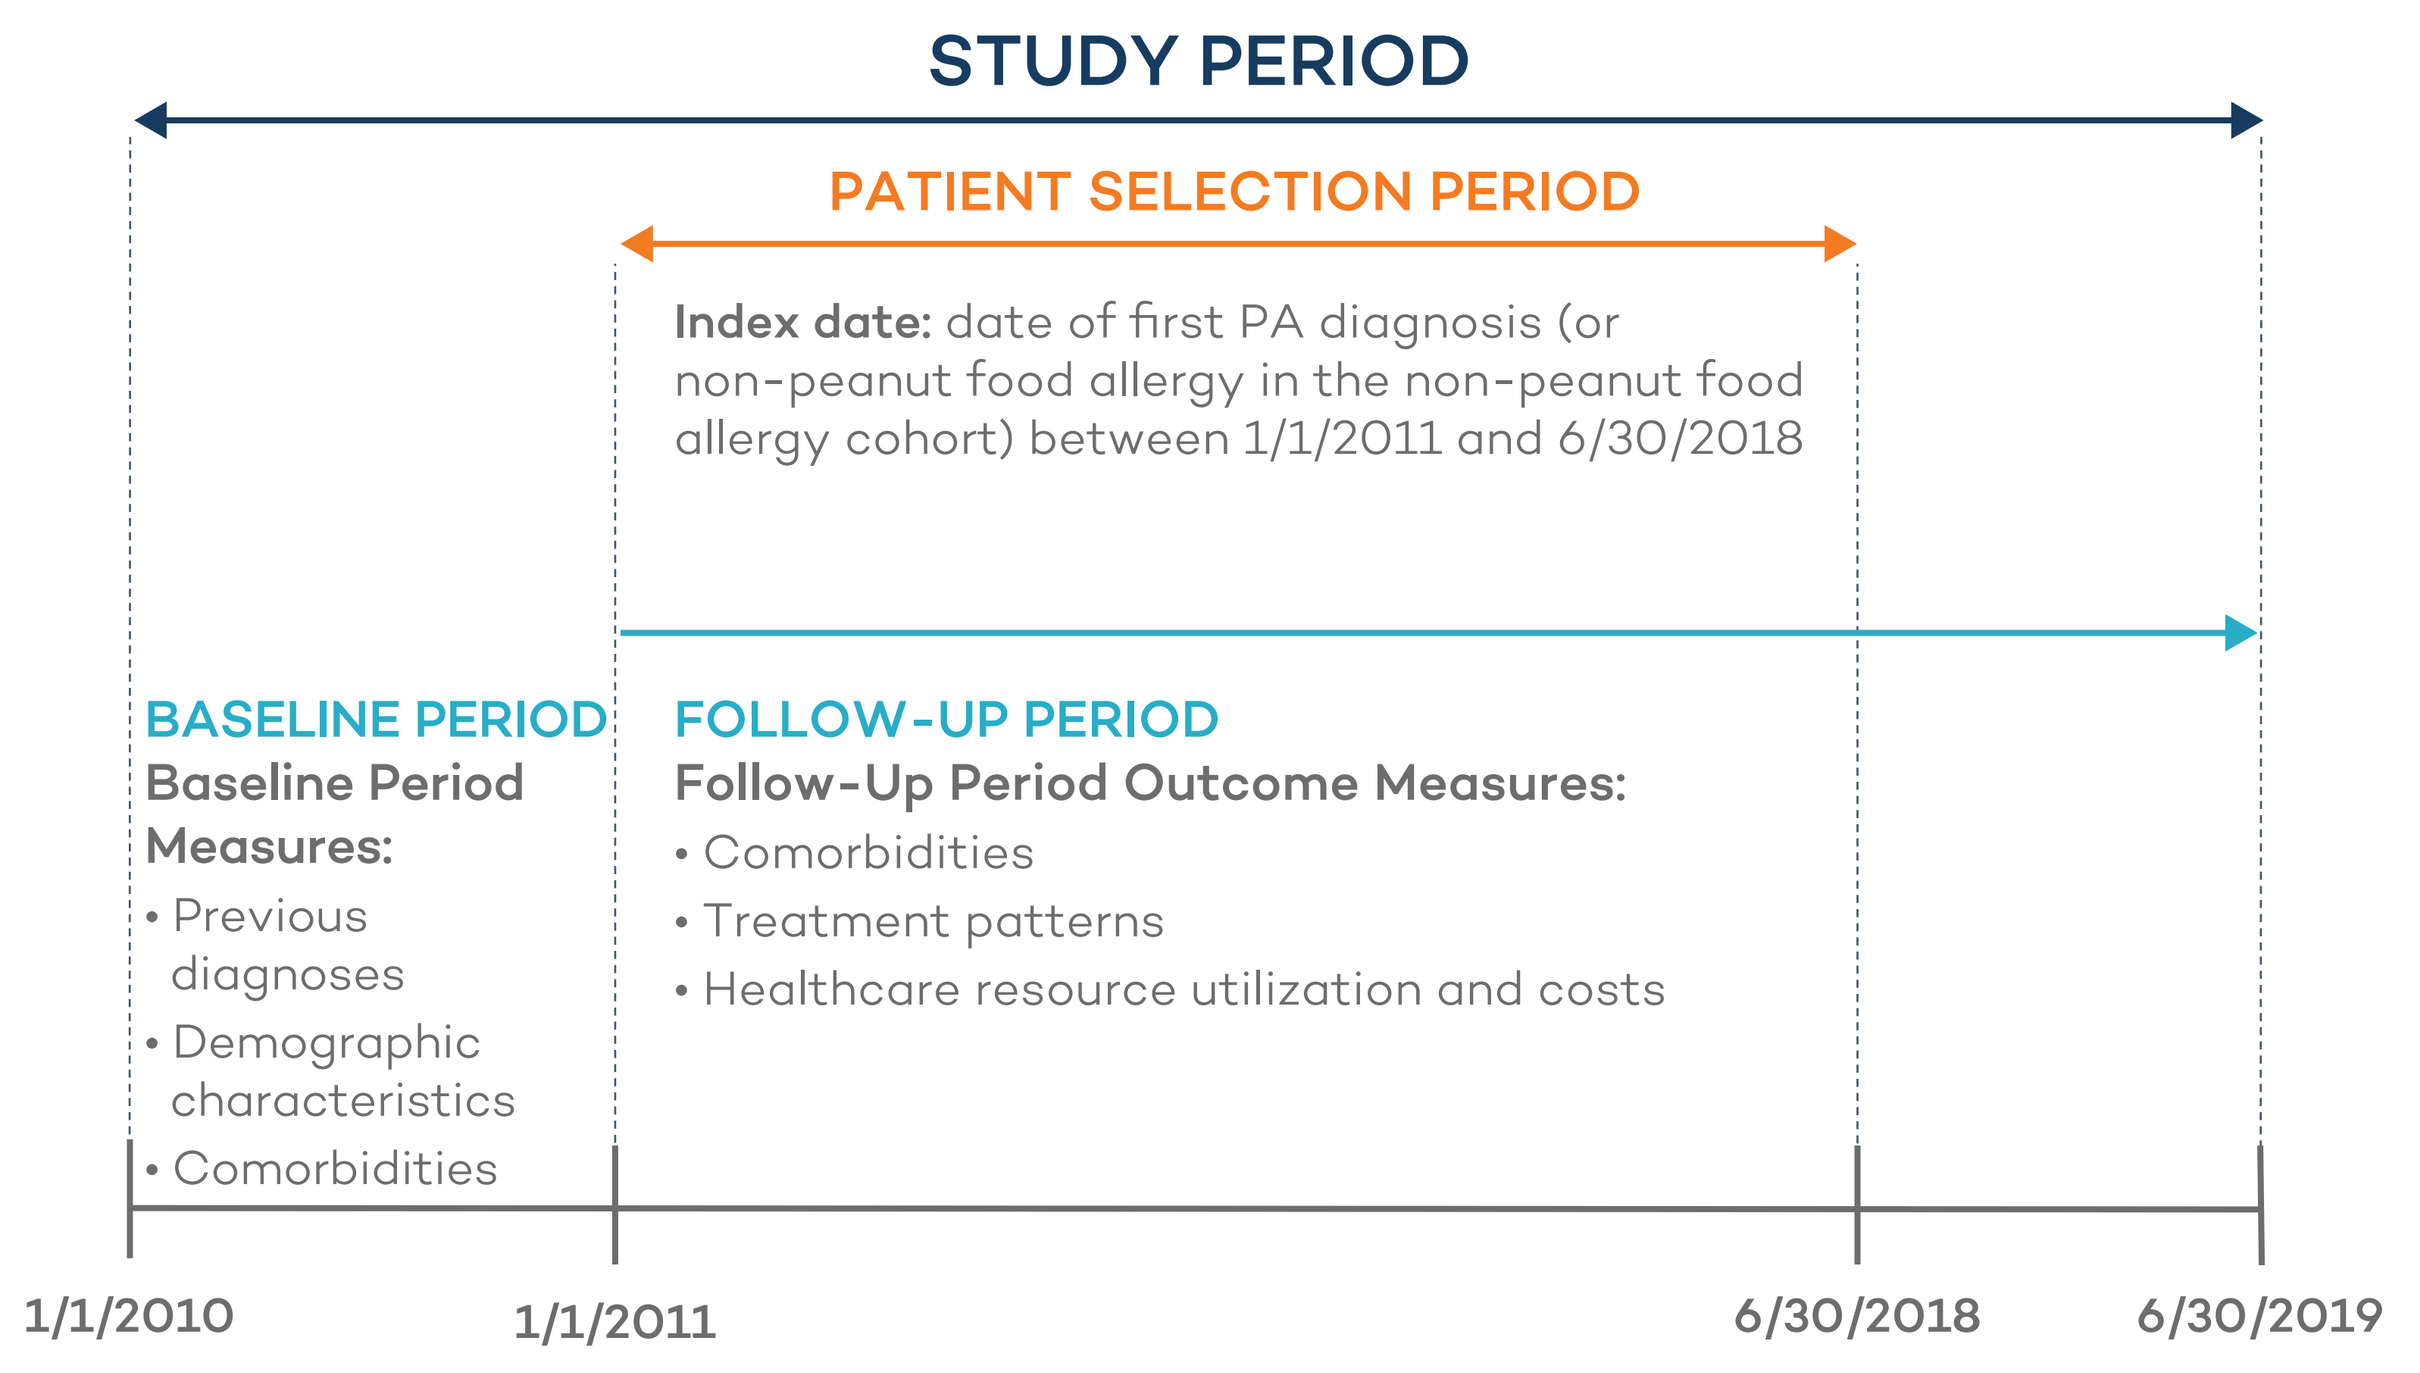
STUDY PERIOD
PATIENT SELECTION PERIOD
Index date: date of first PA diagnosis (or non-peanut food allergy in the non-peanut food allergy cohort) between 1/1/2011 and 6/30/2018
BASELINE PERIOD
Baseline Period Measures:
• Previous diagnoses
• Demographic characteristics
• Comorbidities
FOLLOW-UP PERIOD
Follow-Up Period Outcome Measures:
• Comorbidities
• Treatment patterns
• Healthcare resource utilization and costs
1/1/2010
1/1/2011
6/30/2018
6/30/2019

- Patients with PA or NPAFAC diagnosis/reaction-related codes were identified and included if they had continuous enrollment with medical and pharmacy benefits for at least 12 months before and 12 months after the first PA or NPAFAC diagnosis
- Patients with PA and NPAFAC were matched by gender, age group cohort (0–3, 4–11, 12–18, 19–64, and 0–26), year of index diagnosis, geographic region, and health insurance plan type

Outcomes

- Key study variables assessed are summarized in **Table 1**

Table 1. Summary of Study Variables

DEMOGRAPHICS	CLINICAL CHARACTERISTICS	TREATMENT PATTERNS	HEALTHCARE COSTS AND UTILIZATION
• Total numbers/group (PA and NPAFAC) • Age (number by age cohort) • Sex • Index year • Geographic region • Urban/rural • Insurance plan type	• Number of unique NDC • Number of 3-digit ICD-9-CM and/or ICD-10-CM diagnosis codes	• Allergy-related: o Anaphylactic reactions o Medications o Treatment procedures • Physician specialty	• Inpatient, outpatient, and pharmacy claims associated with: o Any allergy o PA o NPAFAC • Costs associated with specific anaphylactic reaction–related procedures and services

ICD-9-CM/ICD-10-CM=International Classification of Diseases, 9th/10th Revision, Clinical Modification; NDC=National Drug Codes.

Statistical Analyses

- Chi-square tests were used to evaluate statistical significance of differences between groups for categorical variables, and t-tests were used for continuous variables; the threshold P value for statistical significance was 0.05

RESULTS

Patient Demographics and Clinical Characteristics

- A total of 72,854 patients with PA and 166,825 age-matched NPAFAC were included in this analysis (**Table 2**)
 - o Of the patients with PA, 39,068 (53.6%) had ≥1 allergist visit
- Among patients with PA, the mean number of NDC were higher for PA persons with an allergist visit than in those without during both baseline (5.3 vs 4.7; P<0.001) and follow-up (5.7 vs 5.4; P<0.001)
 - o The mean number of ICD-10 codes were also higher for PA persons with an allergist visit versus those without during both baseline (8.9 vs 7.9; P<0.001) and follow-up (10.5 vs 10.2; P<0.001)

Table 2. Patient Attrition

	PATIENTS WITH PA (n=72,854)	NPAFAC (n=166,825)
Age, years		
0–3	21.4%	12.2%
4–11	42.6%	21.5%
12–18	20.1%	14.9%
19–64	16.0%	51.4%
0–26	90.4%	58.3%
Non-peanut food allergies	N/A	
Patients with allergies to multiple foods*		8.3%
Patients with single food allergy only		91.7%
Milk products only		6.2%
Eggs only		5.1%
Seafood only		18.0%
Fruits and vegetables only		1.2%
Tree nuts and seeds only		5.5%
Food additives only		0.2%
Other or unspecified food only		63.7%

*Patients were flagged as having allergies to multiple foods if they had evidence of a combination of claims for 2 or more of the following: milk products, eggs, seafood, fruits and vegetables, tree nuts and seeds, food additives, other or unspecified foods.

Treatment Patterns

- Rates of epinephrine claims (ie, prescriptions filled), mean epinephrine costs, and proportion of patients with peanut anaphylaxis episodes were higher among the PA group with versus without an allergist visit (all P<0.001) (**Table 3**)
 - o The overall proportion of PA patients with an allergist visit who experienced an anaphylaxis episode varied only slightly by age group: 0–3 years, 47%; 4–11 years, 49.8%; and 12–18 years, 49.2%
 - o The proportion of PA patients without an allergist visit who experienced an anaphylaxis episode was highest in the 0–3 years age group (25%) and was lower in the 4–11 years (19.8%) and 12–18 years (18.6%) age groups
 - o PA-persons with an allergist visit versus those without an allergist visit were prescribed an epinephrine autoinjector at a higher rate (69.9% vs 63.3%; RR, 1.67; P<0.001), resulting in higher mean epinephrine costs
- The proportion of patients with an anaphylaxis episode in the prior 12 months was higher in the PA than the NPAFAC group (53.1% vs 31.6%; P<0.001)
- Rates of epinephrine claims for PA patients (66.8%) were higher than nearly all other food allergens, which ranged from 27.9% (milk) to 73.7% (tree nuts) (**Figure 2**)

Table 3. Treatment Patterns in PA Patients With and Without an Allergist Visit

	PA PATIENTS WITH ≥1 ALLERGIST VISIT (n=30,699)	PA PATIENTS WITHOUT ALLERGIST VISIT (n=30,699)	*P* VALUE
Rates of epinephrine claims	69.9%	63.3%	<0.001
Mean epinephrine costs	$676	$493	<0.001
Proportion with peanut anaphylaxis episodes	48.9%	20.7%	<0.001

Figure 2. Rates of Epinephrine Claims by Allergen

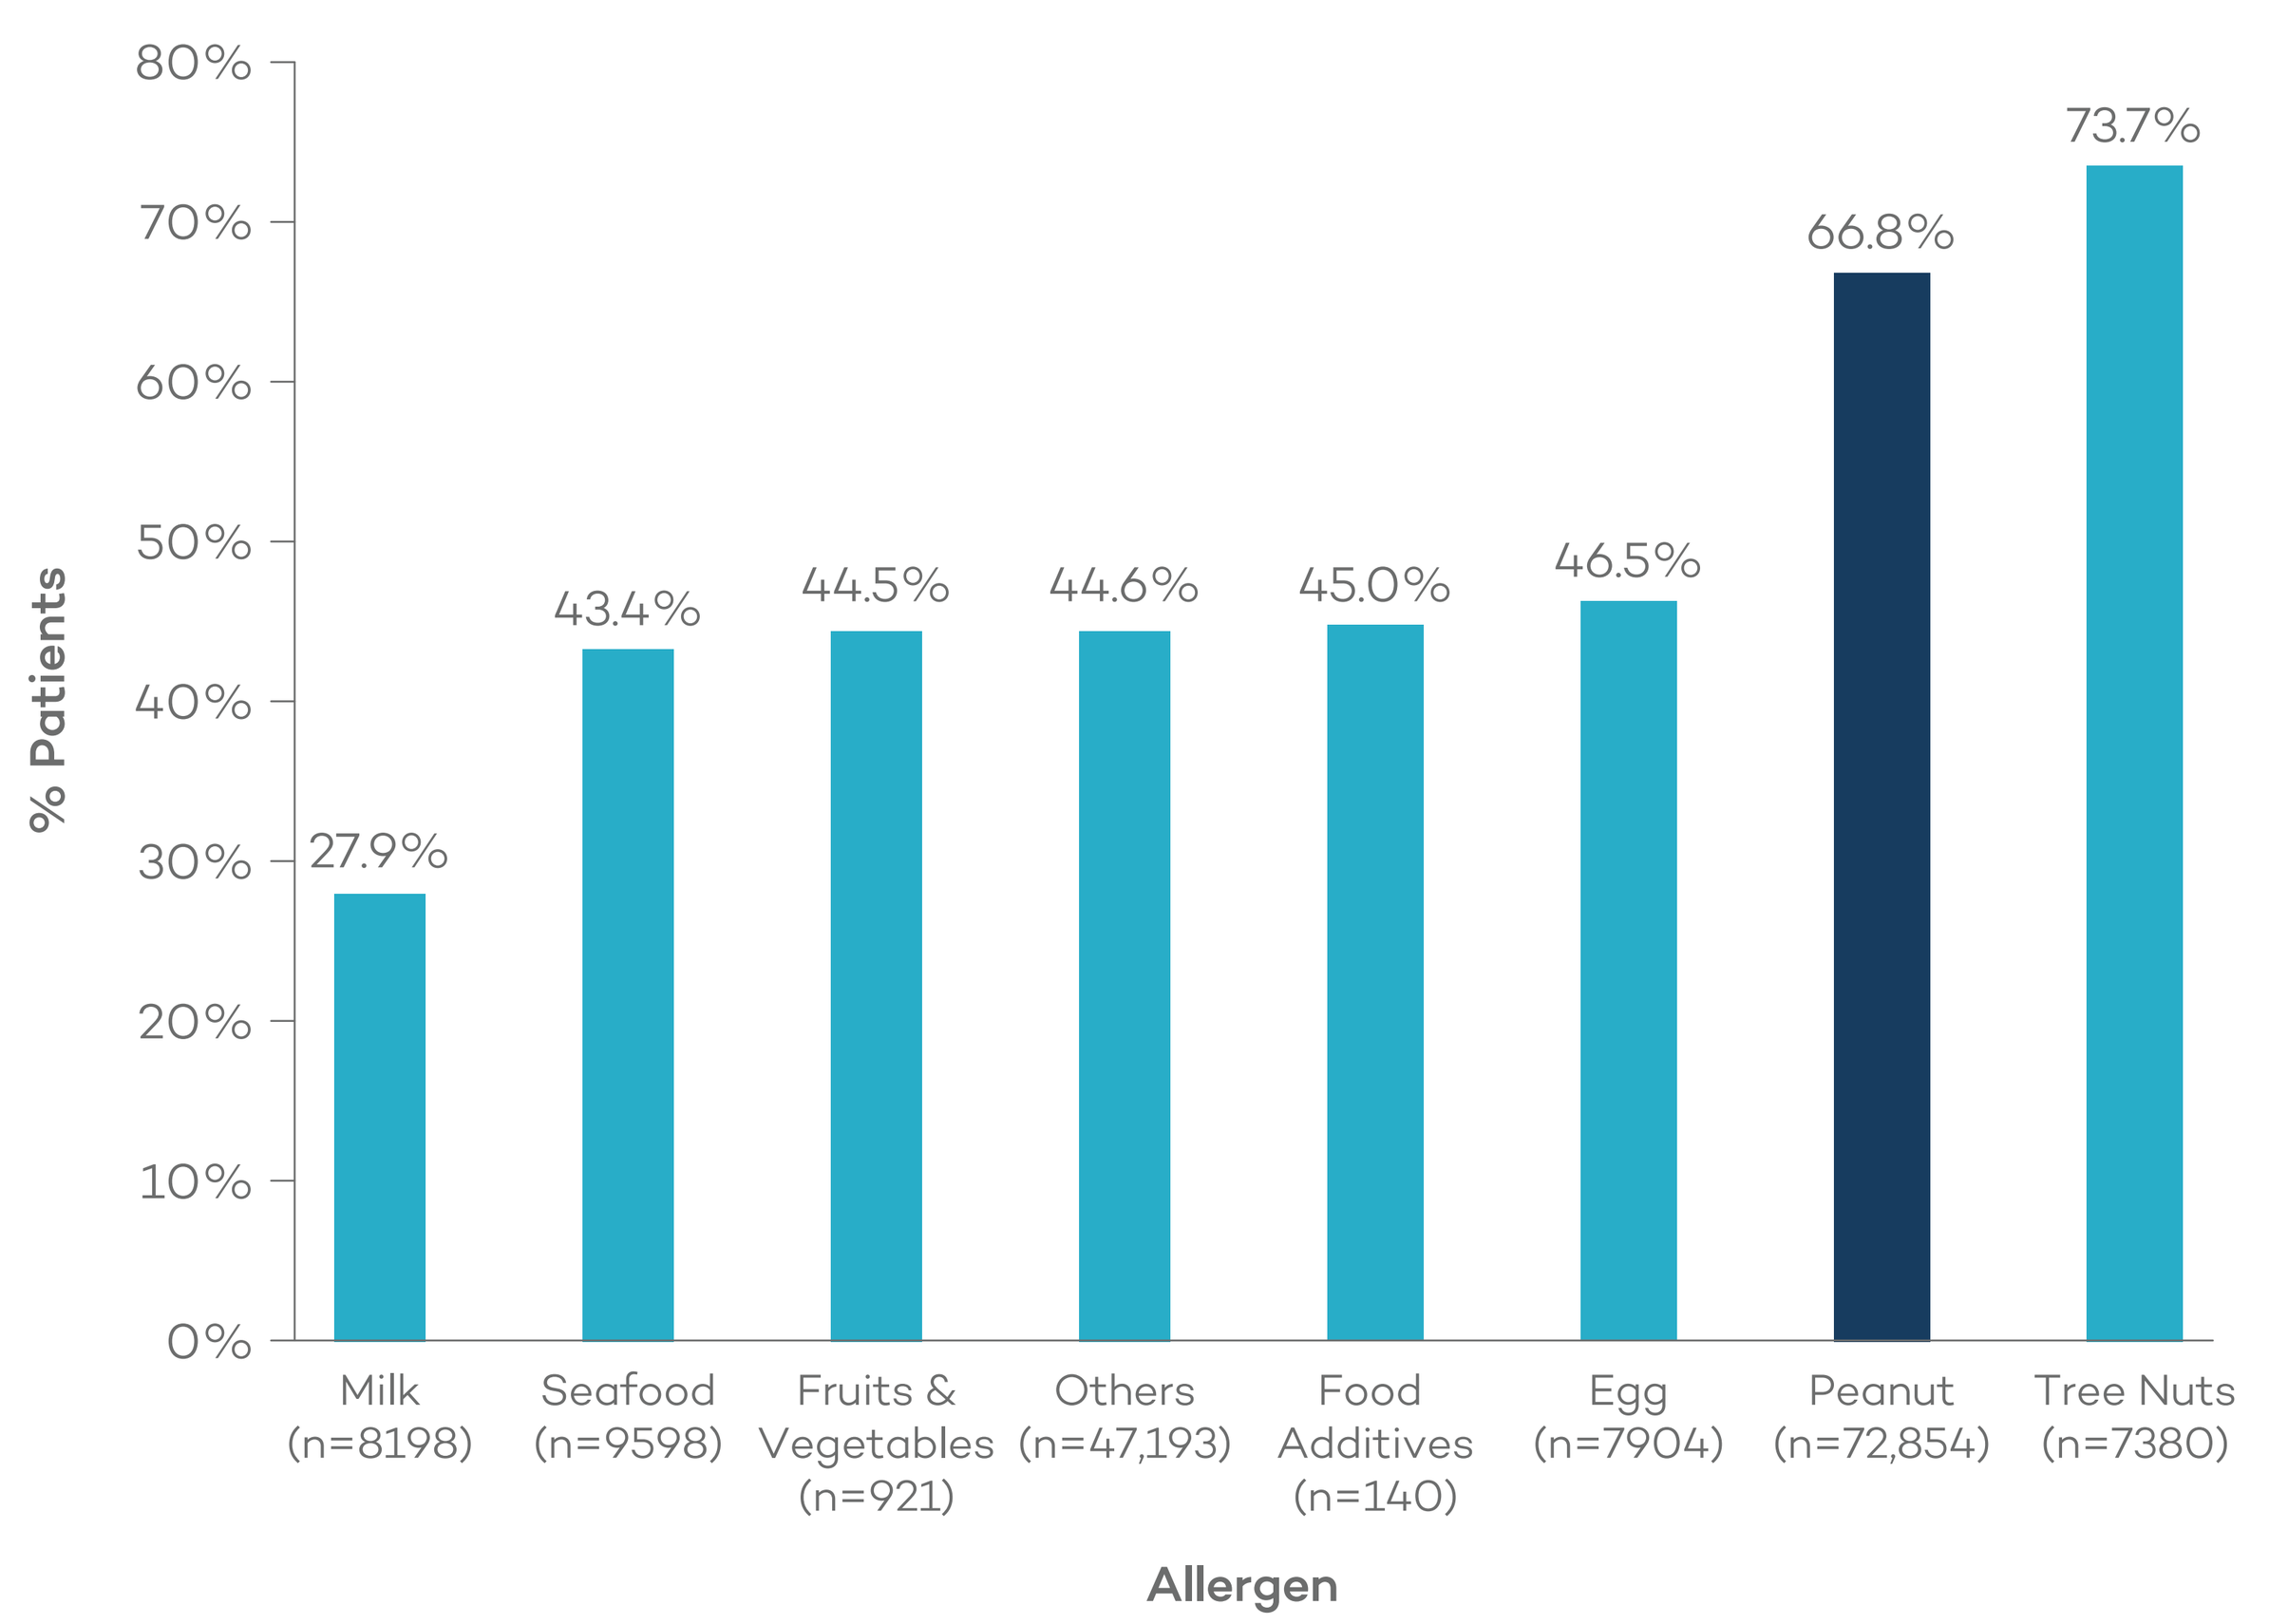
% Patients
80%
70%
60%
50%
40%
30%
20%
10%
0%
27.9%
43.4%
44.5%
44.6%
45.0%
46.5%
66.8%
73.7%
Milk (n=8198)
Seafood (n=9598)
Fruits & Vegetables (n=921)
Others (n=47,193)
Food Additives (n=140)
Egg (n=7904)
Peanut (n=72,854)
Tree Nuts (n=7380)
Allergen

- Mean total healthcare costs were higher in the NPAFAC group than the PA group ($7863 vs $7261; P<0.001) and lower for PA persons with versus without an allergist visit ($6347 vs $8270; P<0.001) (**Figure 3**)
 - o There were no significant differences in PA reaction–related costs between PA groups
 - o Costs for both PA patients and NPAFAC were highest in the 12–18 years age group, and were higher in the 0–3 years than the 4–11 years age group
 - o Mean total healthcare costs by specific food allergen ranged from $6517 (tree nuts only) to $14,424 (seafood only) (**Figure 4**)

Figure 3. Mean Total Healthcare Costs Among Patients With PA (With or Without an Allergist Visit) and NPAFAC

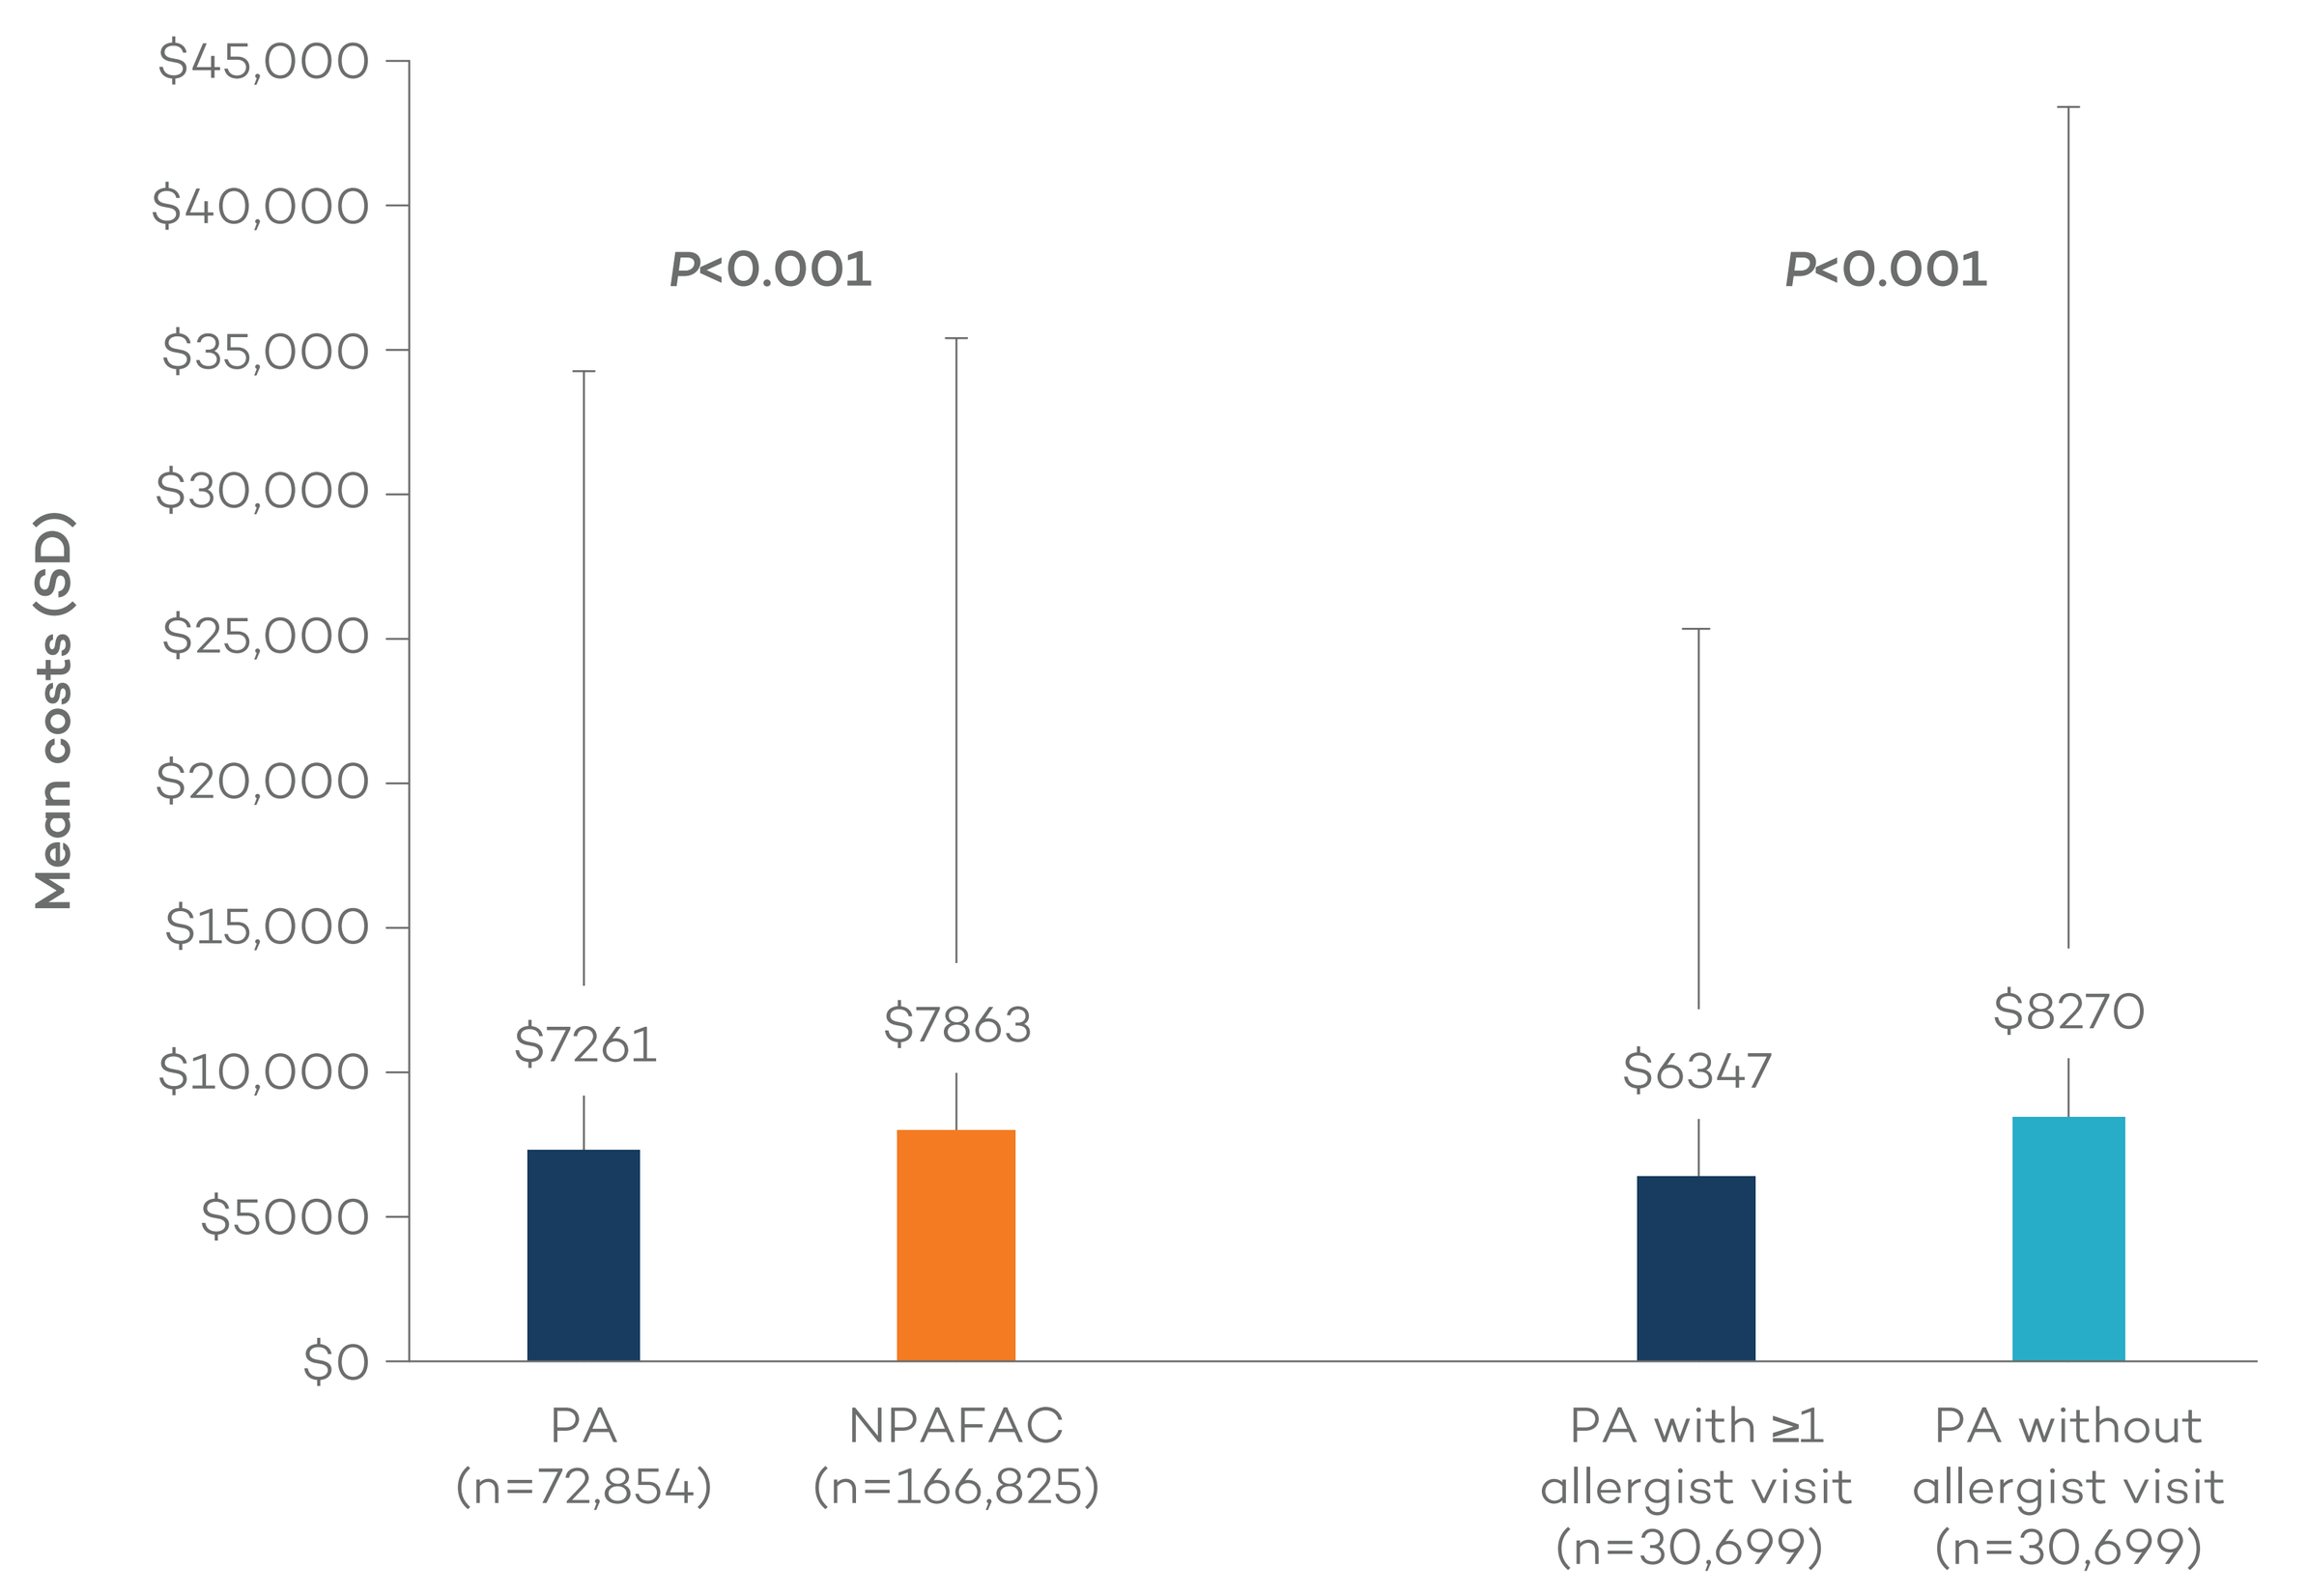
Mean costs (SD)
$45,000
$40,000
$35,000
$30,000
$25,000
$20,000
$15,000
$10,000
$5000
$0
P<0.001
P<0.001
$7261
$7863
$6347
$8270
PA (n=72,854)
NPAFAC (n=166,825)
PA with ≥1 allergist visit (n=30,699)
PA without allergist visit (n=30,699)

SD=standard deviation.

Figure 4. Mean Total Healthcare Costs by Allergen

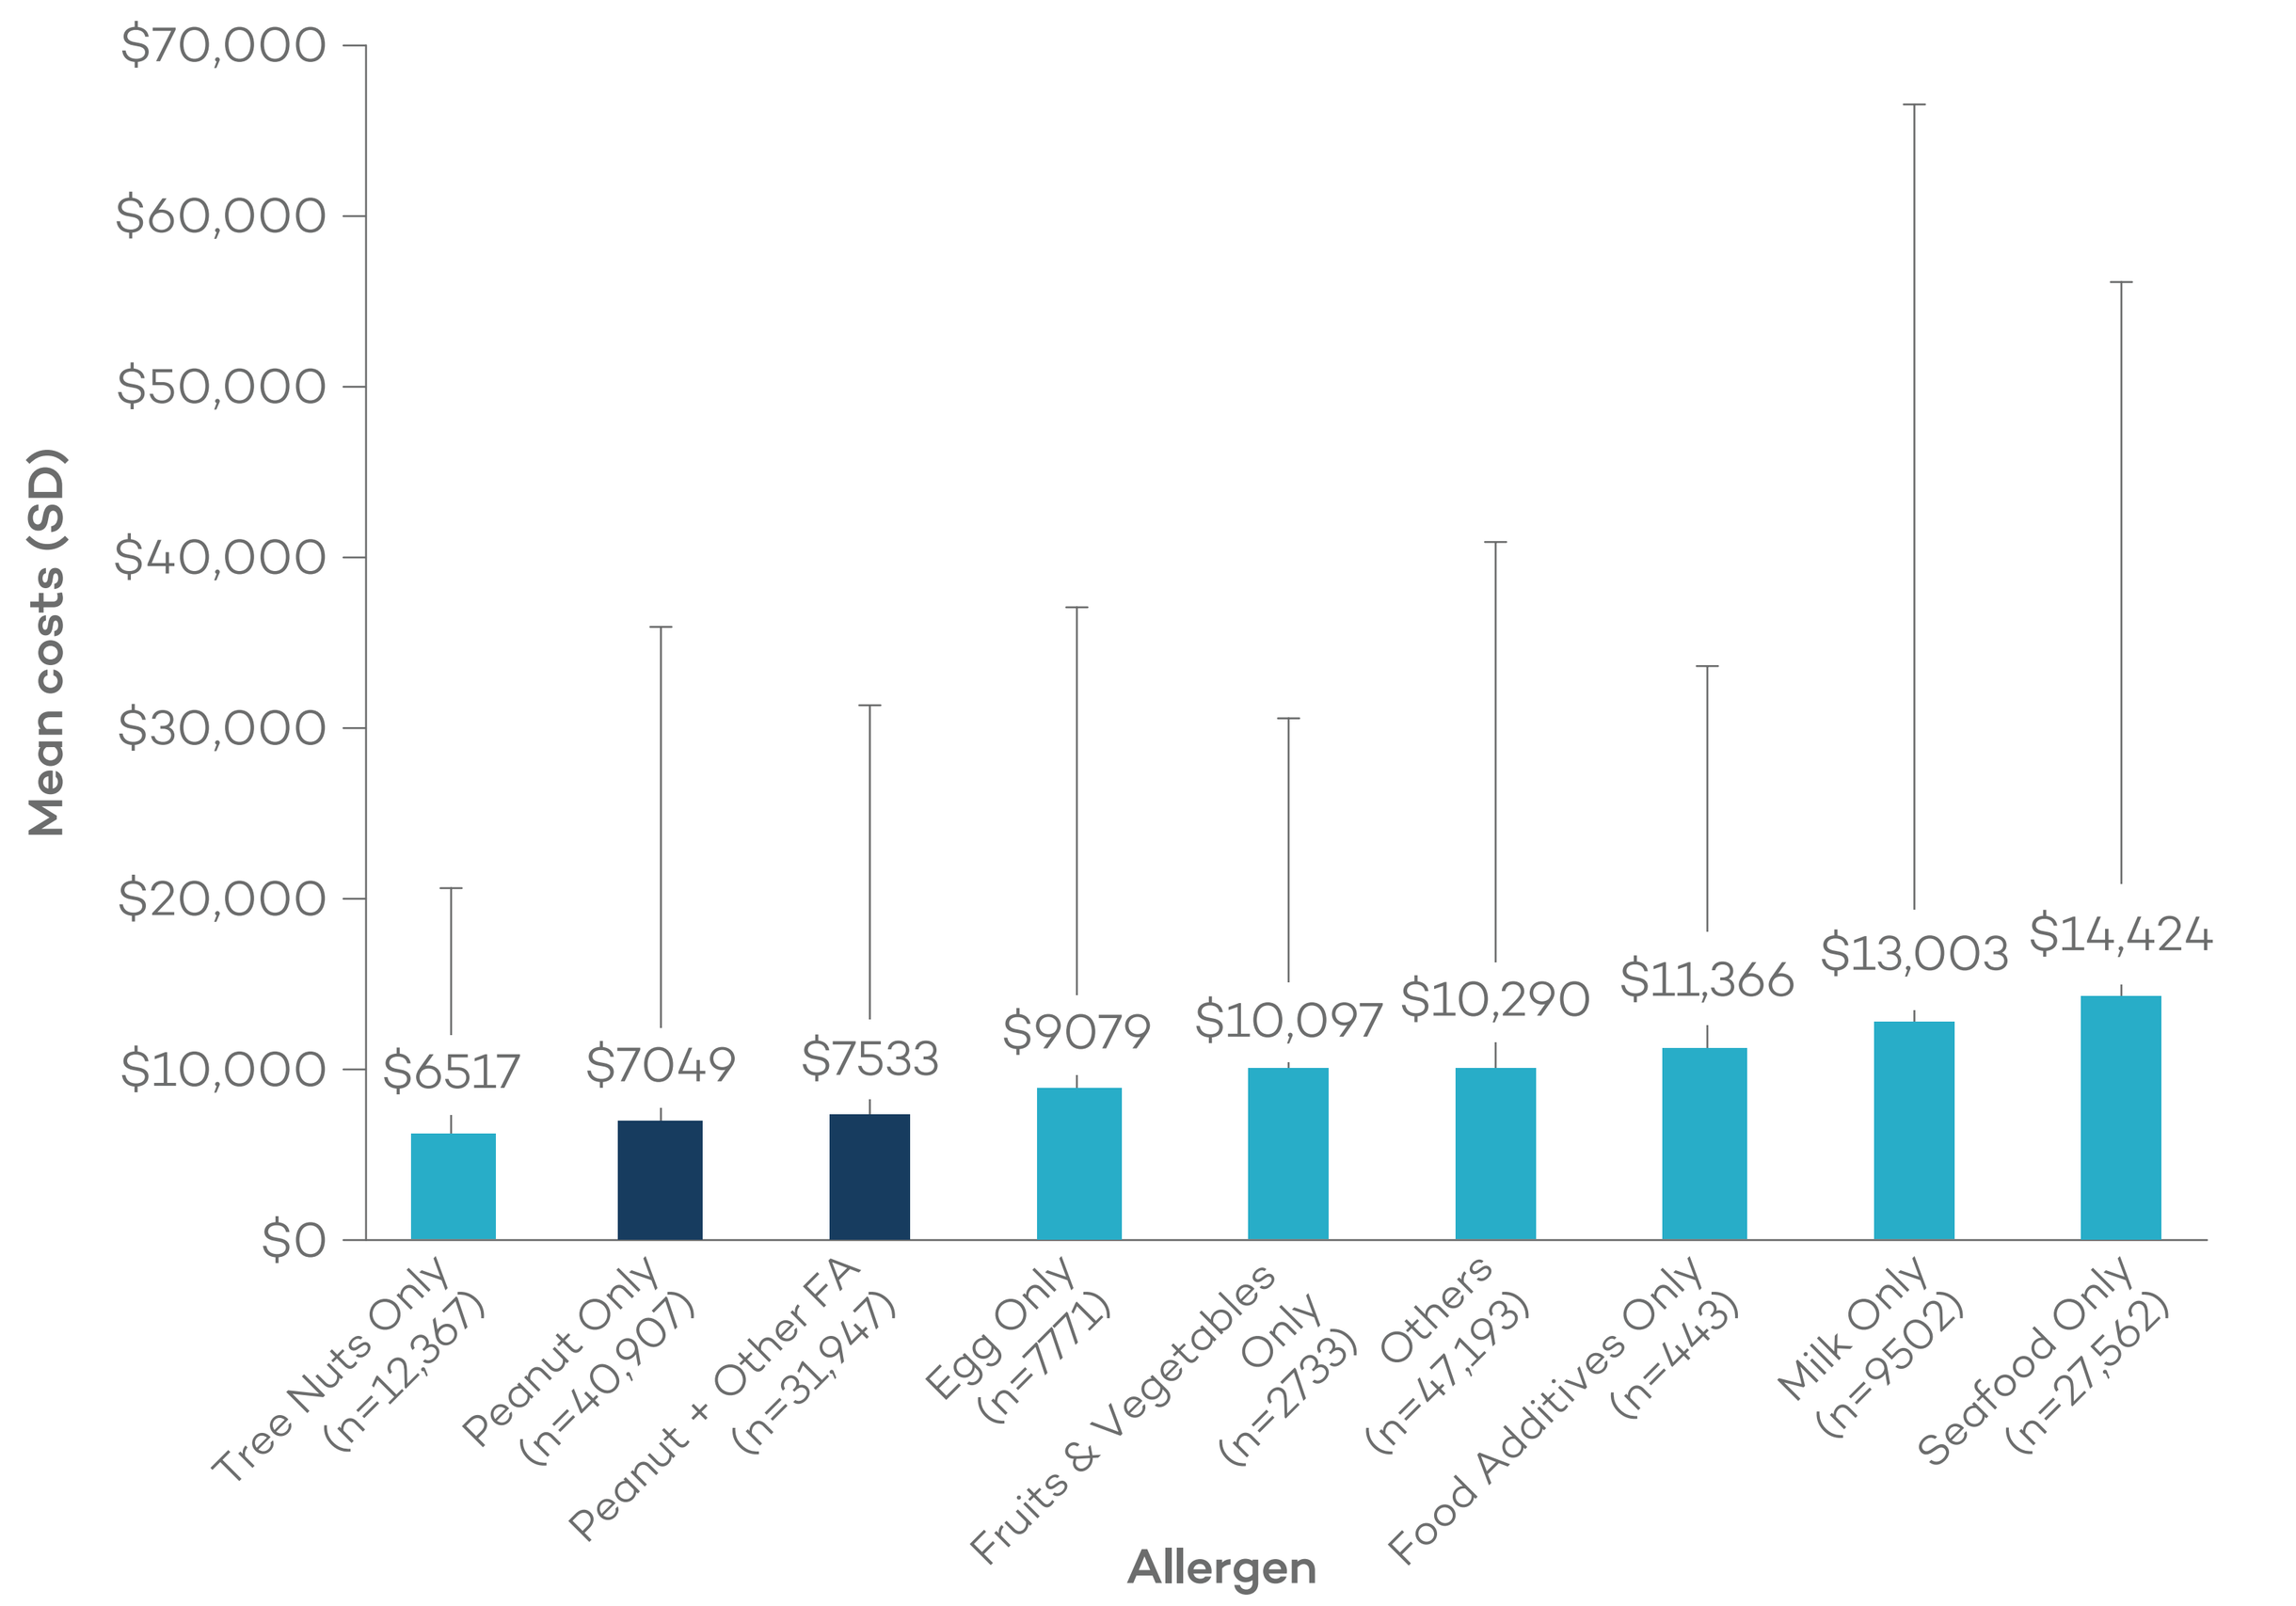
Mean costs (SD)
$70,000
$60,000
$50,000
$40,000
$30,000
$20,000
$10,000
$0
$6517
$7049
$7533
$9079
$10,097
$10,290
$11,366
$13,003
$14,424
Tree Nuts Only (n=12,367)
Peanut Only (n=40,907)
Peanut + Other FA (n=31,947)
Egg Only (n=7771)
Fruits & Vegetables Only (n=2733)
Others (n=47,193)
Food Additives Only (n=443)
Milk Only (n=9502)
Seafood Only (n=27,562)
Allergen

SD=standard deviation.

CONCLUSIONS

- This study shows that there were higher rates of anaphylaxis among the PA group compared with the NPAFAC group (53.1% vs 31.6%; P<0.001)
- The role of an allergist in the care of someone with PA is highly important. Anaphylaxis claims and epinephrine prescription rates were higher for persons with PA that had an allergist visit versus persons with PA without an allergist visit. This likely reflects that patients with a history of more severe peanut allergy are being managed by an allergist rather than a primary care provider
- Total mean healthcare costs were higher in the NPAFAC group than the PA group, and were lower for PA persons with an allergist visit versus those without an allergist visit
- These findings may suggest that total healthcare costs of PA are lower in patients under the care of an allergist, despite the fact that these patients may have a more severe history as reflected by higher rates of anaphylaxis and epinephrine prescriptions

REFERENCES

1. Shaker M, Chalil JM, Tran O, et al. Commercial claims costs related to health care resource use associated with a diagnosis of peanut allergy. *Ann Allergy Asthma Immunol*. 2020;124(4):357-365.e.1. **2.** McCann W, Hass SL, Norrett K, et al. The peanut allergy burden study: real-world impact of peanut allergy on resource utilization and productivity. *World Allergy Organ J*. 2021;14(3):100525.

ACKNOWLEDGMENTS

This study was sponsored by DBV Technologies. Editorial support for the preparation of this poster was provided by International Meetings & Science (IMsci), funded by DBV Technologies.

American Academy of Allergy, Asthma & Immunology Annual Meeting
Phoenix, AZ; February 25–28, 2022