VITESSE Phase 3 Trial: Efficacy and Safety of Epicutaneous Immunotherapy in Children With Peanut Allergy Aged 4 Through 7 Years

David M. Fleischer, MD;[1] Douglas P. Mack, MD;[2] Julie Wang, MD;[3] Jeffrey Leflein, MD;[4] Michael O'Sullivan, MD;[5] Anne-Sophie Chhim, MD;[6] Katharine J. Bee, PhD;[6] Hugh A. Sampson, MD[3]

[1]Children's Hospital Colorado, University of Colorado, Aurora, CO, USA; [2]McMaster University, Hamilton, ON, Canada; [3]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [4]Allergy and Immunology Associates of Ann Arbor, PC, Ann Arbor, MI, USA; [5]Perth Children's Hospital, The Kids Research Institute Australia, Nedlands, Western Australia, Australia; [6]DBV Technologies SA, Châtillon, France

RATIONALE

- Epicutaneous immunotherapy (EPIT) is an innovative approach to food allergy treatment
- The VIASKIN® Peanut Patch utilizes EPIT to provide a non-invasive treatment that uses the immune properties of the skin to induce desensitization while limiting systemic allergen exposure
- This innovative approach involves the administration of a peanut patch containing 250 µg of peanut protein to intact skin to induce desensitization (**Figure 1**)[1-5]
- The VIASKIN® Peanut Patch is an investigational treatment for children with peanut allergy aged 4 through 7 years, and has not yet been approved by the US Food and Drug Administration

Figure 1. VIASKIN® Peanut Patch for Children Aged 4-7 Years

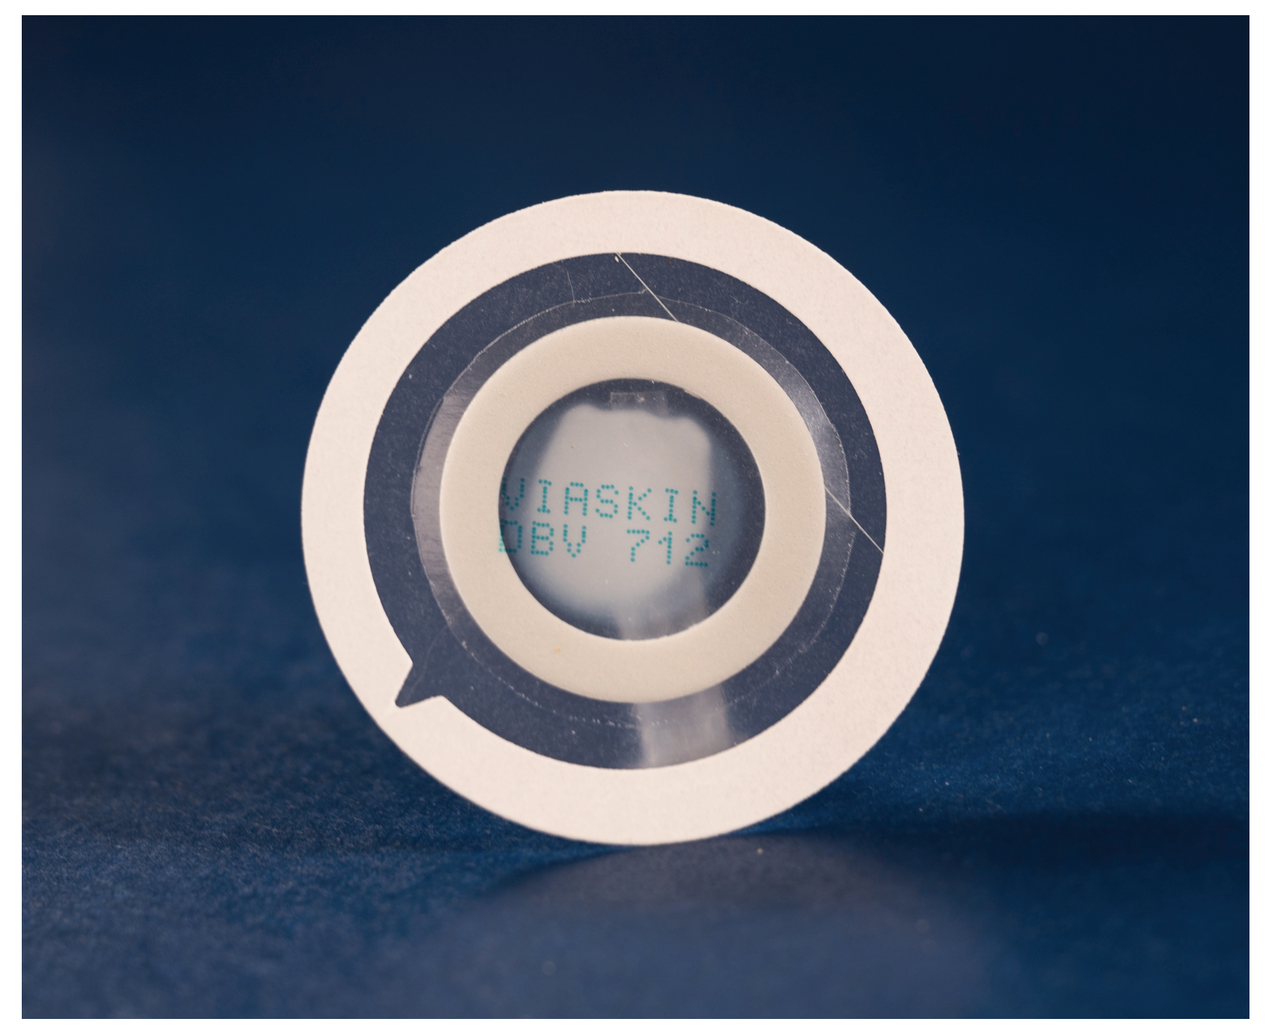
VIASKIN
DBV 712

The VIASKIN® Peanut Patch is 44 mm in diameter.

OBJECTIVE

- To report the primary efficacy and safety of the VIASKIN® Peanut Patch in children aged 4 through 7 years in the double-blind, placebo-controlled (DBPC) Phase 3 VITESSE trial (NCT05741476)

METHODS

- Participants were randomized 2:1 to the VIASKIN® Peanut Patch or placebo for 12 months, with the option of active treatment for up to 36 months in an ongoing open-label extension (**Figure 2**)
- Eligibility criteria included peanut-specific immunoglobulin E (IgE) >0.7 kU_A/L, peanut skin prick test ≥6 mm, and eliciting dose (ED) of ≤100 mg of peanut protein at DBPC food challenge (DBPCFC)

Figure 2. VITESSE Study Design

Phase 3 global trial (NCT05741476)

- Children with peanut allergy (aged 4 through 7 years)
- 87 sites in Australia, Canada, Europe, and the USA
- Key inclusion criteria:
 - Baseline ED ≤100 mg
 - psIgE >0.7 kU_A/L
 - Skin prick test ≥6 mm

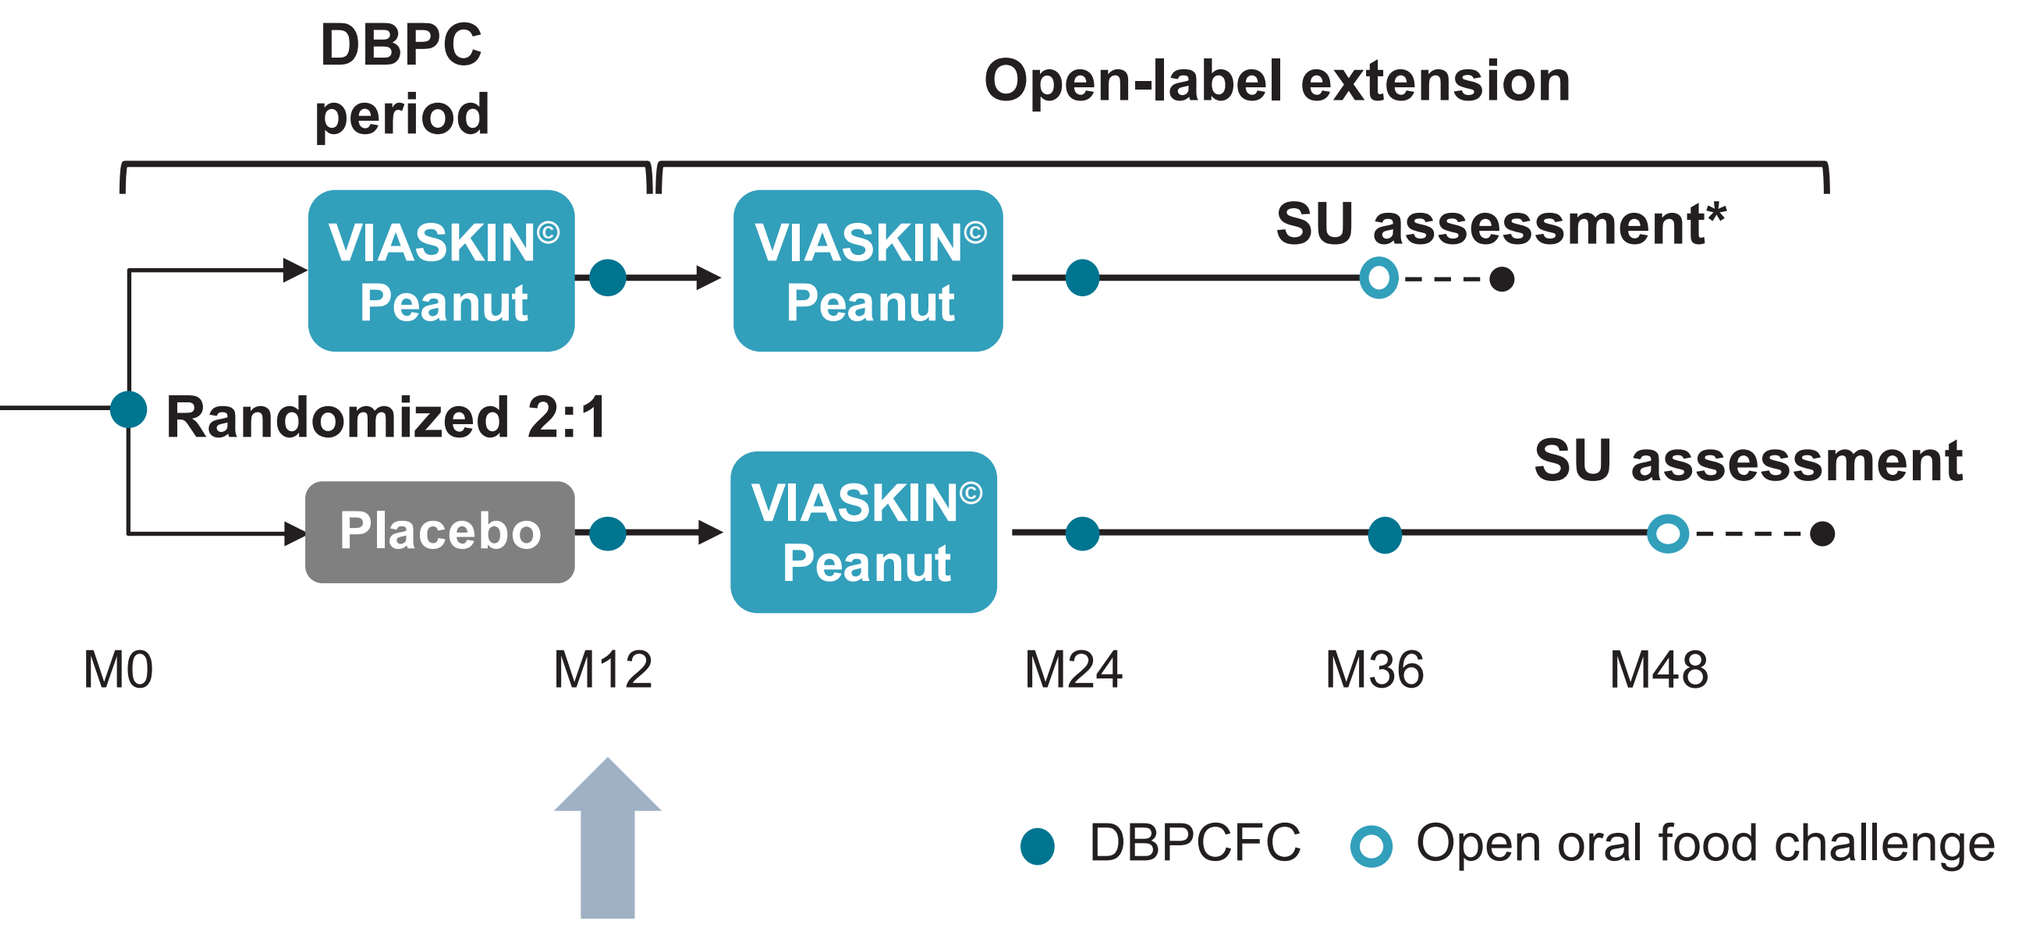
DBPC period
Open-label extension
VIASKIN© Peanut
VIASKIN© Peanut
SU assessment*
Randomized 2:1
Placebo
VIASKIN© Peanut
SU assessment
M0
M12
M24
M36
M48
DBPCFC
Open oral food challenge

*SU assessment involves an open oral food challenge every 2 months for 6 months after stopping treatment in eligible participants.
DBPC, double-blind, placebo-controlled; DBPCFC, double-blind, placebo-controlled food challenge; ED, eliciting dose; M, month; psIgE, peanut-specific immunoglobulin E; SU, sustained unresponsiveness.

- The primary endpoint was the difference in the proportion of responders between the VIASKIN® Peanut Patch and placebo groups at month 12, defined as DBPCFC month 12 ED ≥300 mg if baseline ED is ≤30 mg or month 12 ED ≥600 mg if baseline ED is 100 mg
- The average daily wear time was calculated based on time of patch application and removal, as recorded daily in a participant eDiary
- Safety assessments included overall adverse events, local patch application-site reactions, and systemic allergic reactions

REFERENCES: **1.** Fleischer DM, et al. *JAMA*. 2019;321(10):946-955; **2.** Fleischer DM, et al. *J Allergy Clin Immunol*. 2020;146(4):863-874; **3.** Pongracic JA, et al. *J Allergy Clin Immunol Pract*. 2022;10(7):1864-1873.e10; **4.** Wang J, Sampson HA. *Pediatr Allergy Immunol.* 2018;29(4): 341-349; **5.** Greenhawt M, et al. *N Engl J Med*. 2023;388(19):1755-1766.

FUNDING SOURCE/ACKNOWLEDGMENTS: The VITESSE trial was sponsored by DBV Technologies. Editorial support for the preparation of this poster was provided by LiNK Health Group, funded by DBV Technologies.

VIASKIN® Peanut Patch is an investigational agent and has not yet been approved by the US Food and Drug Administration or any other regulatory authority. 2026, DBV Technologies. All rights reserved.

Presented at the Eastern Allergy Conference, May 28-31, 2026; Palm Beach, Florida, USA

KEY POINTS

- Efficacy results from VITESSE, the largest food allergy immunotherapy trial to date, demonstrated that 12 months of EPIT with the VIASKIN® Peanut Patch resulted in significantly greater responder rates vs placebo (46.6% vs 14.8%, respectively; p<0.0001) in a highly sensitized population of children with peanut allergy aged 4 through 7 years
- Average daily wear time was high (>23 hours) in both study groups, indicating that participants applied the VIASKIN® Peanut Patch as intended and designed for daily use
- The safety profile of the VIASKIN® Peanut Patch was favorable; most TEAEs were mild local skin reactions and there were low rates of treatment-related anaphylaxis and epinephrine use
- The low rate of discontinuation due to TEAEs and high rate of compliance suggest that the VIASKIN® Peanut Patch was well tolerated by participants

RESULTS

- 654 participants were randomized in the VITESSE trial (**Figure 3**)

Figure 3. VITESSE Consort Flow Diagram

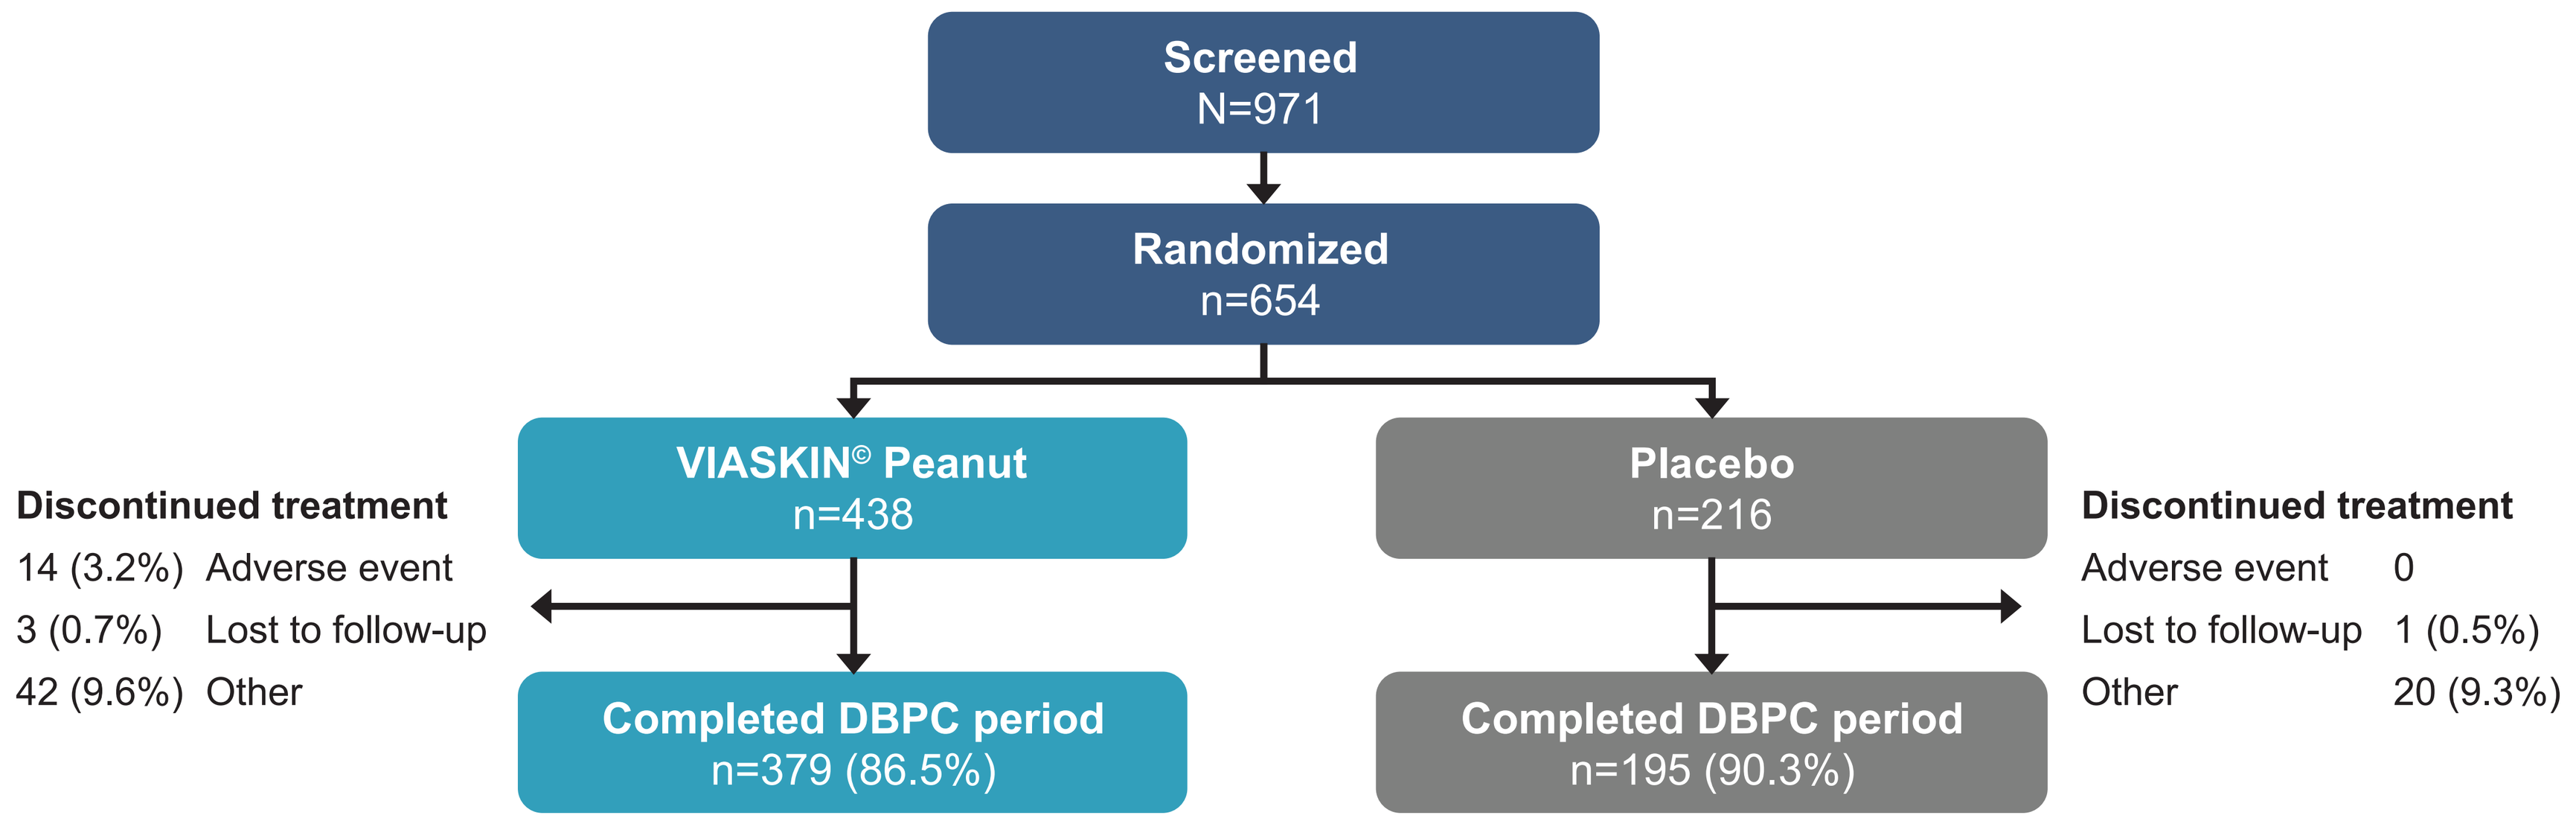
Screened
N=971
Randomized
n=654
VIASKIN© Peanut
n=438
Placebo
n=216
Discontinued treatment
14 (3.2%) Adverse event
3 (0.7%) Lost to follow-up
42 (9.6%) Other
Discontinued treatment
Adverse event 0
Lost to follow-up 1 (0.5%)
Other 20 (9.3%)
Completed DBPC period
n=379 (86.5%)
Completed DBPC period
n=195 (90.3%)

DBPC, double-blind, placebo-controlled.

- Participant demographics and baseline characteristics were well balanced between study groups and treatment compliance was high in both study groups (**Table 1**)
- There was a significantly greater responder rate with the VIASKIN® Peanut Patch vs placebo (46.6% vs 14.8%, respectively; p<0.0001; Δ=31.8% [95% confidence interval: 24.5, 39.0]) (**Figure 4**)

Table 1. Baseline Demographics

	VIASKIN© Peanut (n=438)	Placebo (n=216)
Age, years, median (Q1, Q3)	5.7 (4.8, 6.9)	5.7 (4.8, 6.7)
Sex, n (%)		
Male	265 (60.5)	142 (65.7)
Female	172 (39.3)	74 (34.3)
Undifferentiated	1 (0.2)	0 (0.0)
Race, n (%)		
White	270 (61.6)	146 (67.6)
Asian	70 (16.0)	35 (16.2)
Black	22 (5.0)	8 (3.7)
Native Hawaiian or Other Pacific Islander	2 (0.5)	0 (0.0)
Other	61 (13.9)	20 (9.3)
Unknown	13 (3.0)	7 (3.2)
Peanut-specific IgE, kU_A/L		
Median (IQR)	38.0 (11.1, 161.0)	45.3 (10.9, 185.0)
Peanut-specific IgG4, mg/L		
Median (IQR)	0.9 (0.3, 2.2)	0.8 (0.3, 1.9)
Skin prick test, median wheal diameter, mm	11.0	10.8
Medical history (ongoing at baseline), n(%)		
Atopic dermatitis / eczema	269 (61.4)	135 (62.5)
Asthma	155 (35.4)	79 (36.6)
Other food allergy	247 (56.4)	123 (56.9)
Mean compliance, %*	96.1	96.5

*Overall compliance (%) over the study period is defined as the number of systems dispensed minus the number of systems returned divided by overall exposure duration (days) × 100.
Ig, immunoglobulin; IQR, interquartile range; Q, quartile.

- Most treatment-emergent adverse event (TEAEs) were mild local application-site reactions, consistent with previous VIASKIN® Peanut Patch studies (**Table 2**). There were low rates of discontinuations due to TEAEs

Figure 4. Treatment Responders* at Month 12 (ITT Population)

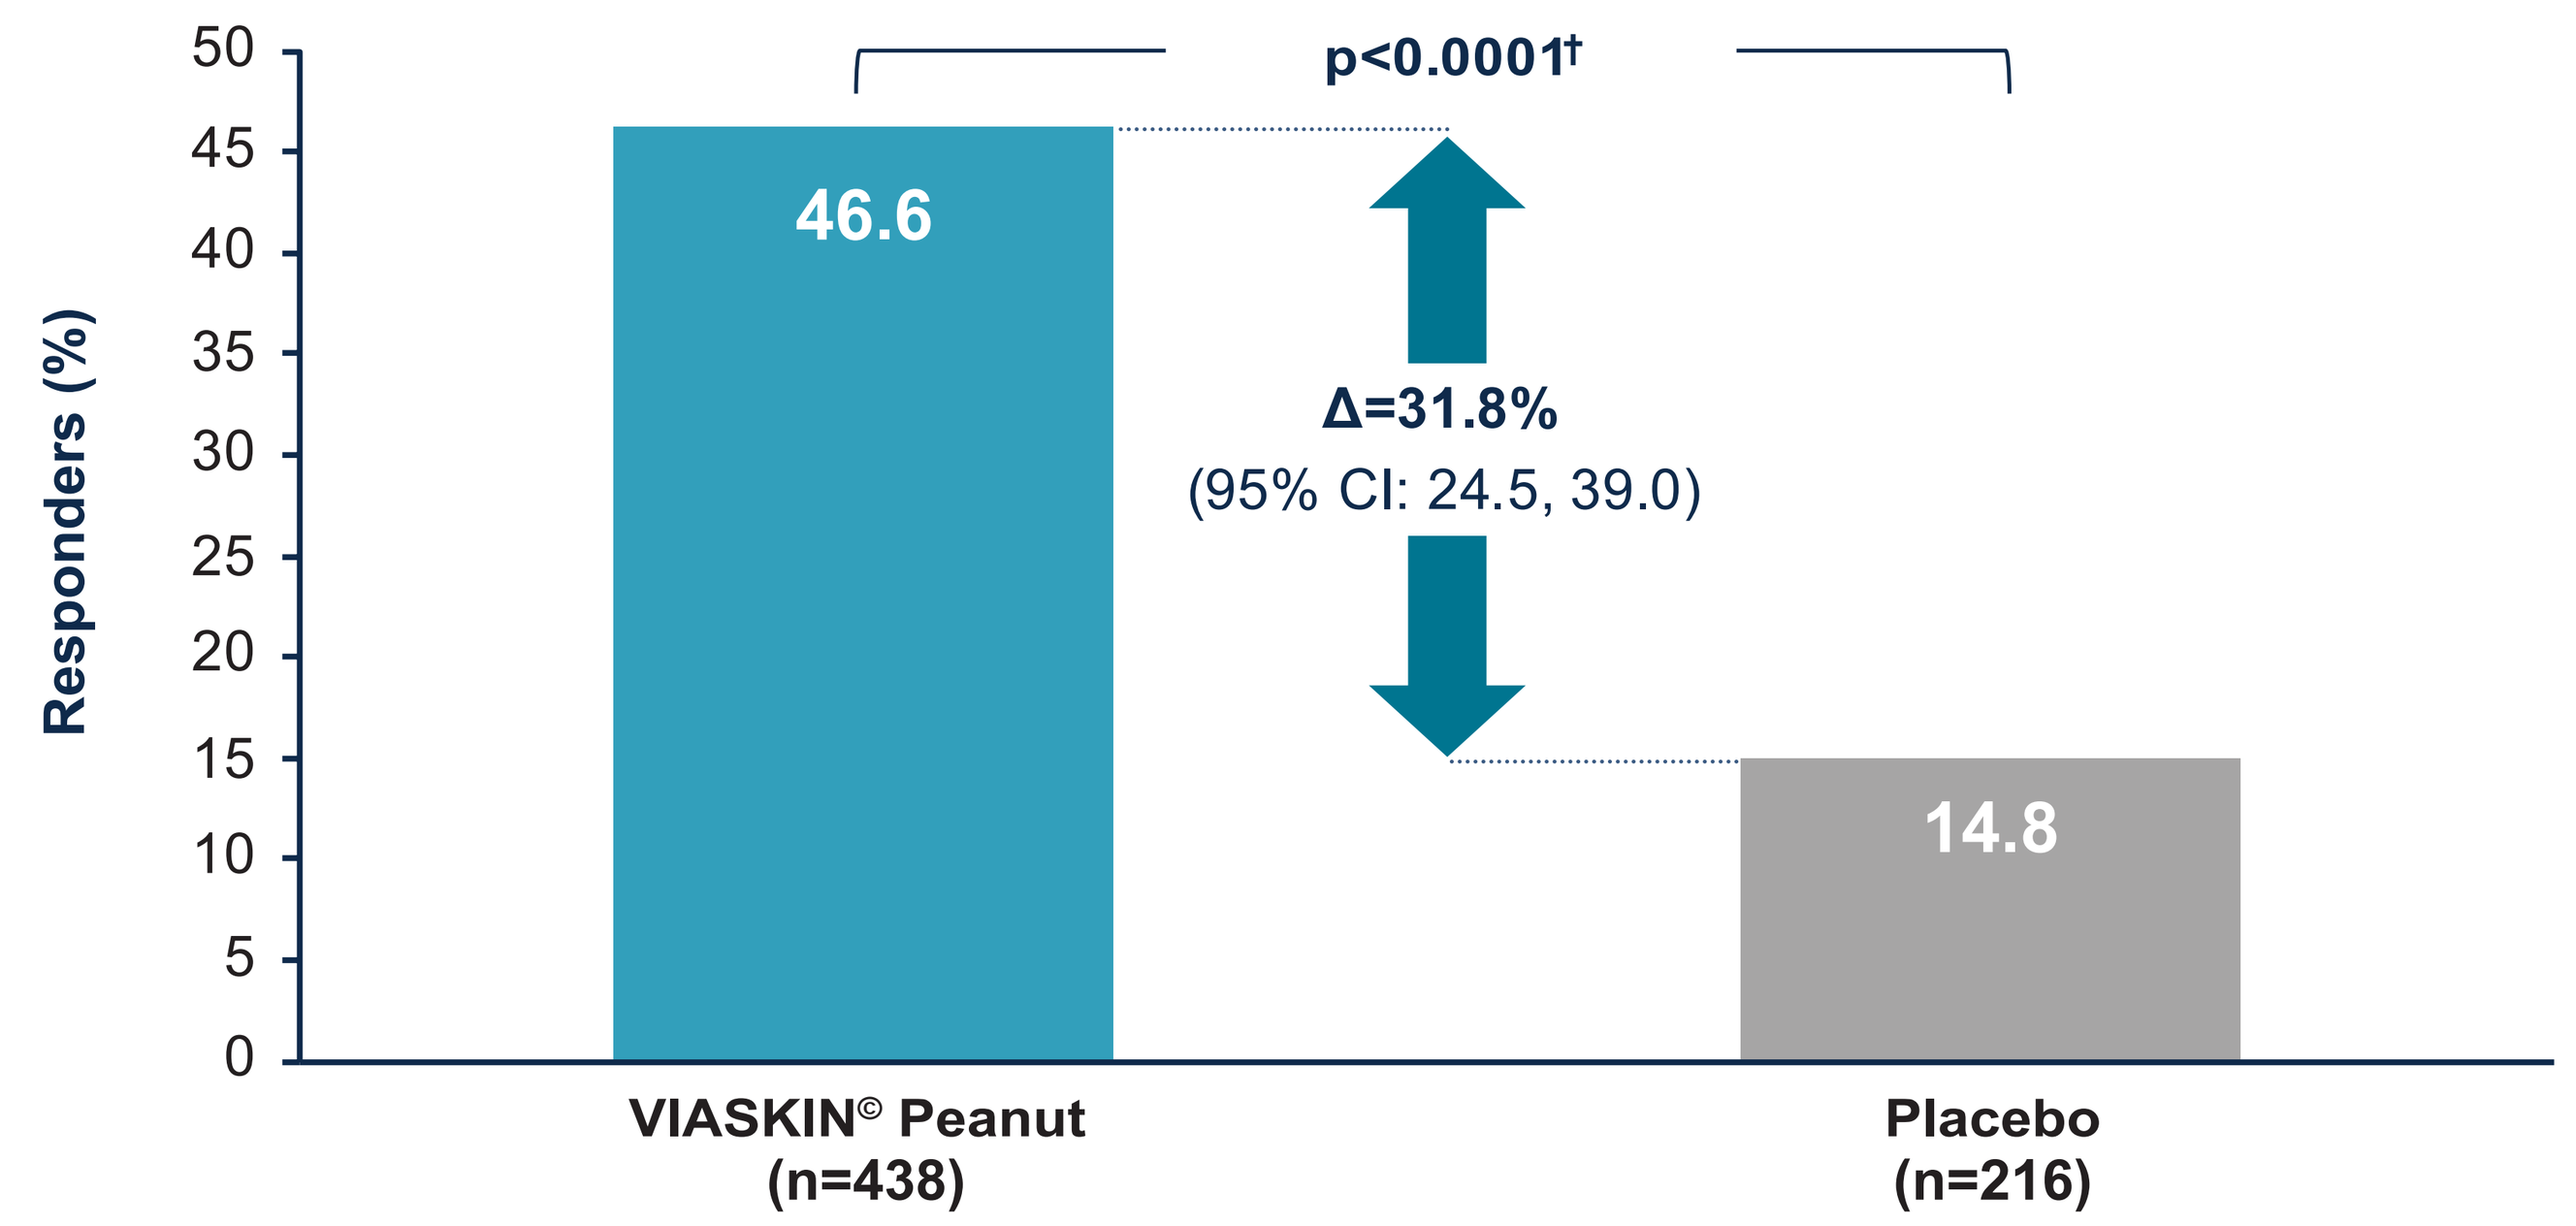
Responders (%)
50
45
40
35
30
25
20
15
10
5
0
p<0.0001†
46.6
Δ=31.8%
(95% CI: 24.5, 39.0)
14.8
VIASKIN© Peanut
(n=438)
Placebo
(n=216)

*Responders were defined as children with a baseline ED ≤30 mg who achieved an ED of ≥300 mg of peanut protein at month 12, or children with a baseline ED of 100 mg who achieved an ED ≥600 mg of peanut protein at month 12. †$p=10^{-17}$.
CI, confidence interval; ED, eliciting dose; ITT, intention-to-treat.

- There were also low rates of treatment-related anaphylaxis and epinephrine use (**Table 2**)
- The mean average daily wear time of the VIASKIN® Peanut Patch was greater than 23 hours and similar to the mean average daily wear time of the placebo patch (**Table 3**)

Table 2. Safety

	VIASKIN© Peanut (n=438)	Placebo (n=216)
TEAEs, number of participants (%)		
Overall	431 (98.4)	206 (95.4)
Mild	420 (95.9)	197 (91.2)
Moderate	231 (52.7)	82 (38.0)
Severe	5 (1.1)	6 (2.8)
Serious	6 (1.4)	5 (2.3)
Treatment-related TEAEs, number of participants (%)		
Overall	404 (92.2)	132 (61.1)
Local	398 (90.9)	127 (58.8)
Severe	2 (0.5)	0 (0.0)
Serious	0 (0.0)	0 (0.0)
Treatment-related anaphylactic reaction	2 (0.5)	0 (0.0)
Treatment-related TEAEs leading to epinephrine use	3 (0.7)	1 (0.5)
TEAEs leading to permanent study discontinuation	14 (3.2)	0 (0.0)

TEAE, treatment-emergent adverse event.

Table 3. Median Average Daily Wear Time

	Global diary (daily) safety set†		Global diary (daily) per protocol†‡	
	VIASKIN© Peanut (n=431)	Placebo (n=214)	VIASKIN© Peanut (n=428)	Placebo (n=214)
ADWT* hours, median	24.0	24.8	24.8	25.6

*ADWT is based on the number of participants, not VIASKIN© Peanut Patches. †The daily reporting allowed for a 4-hour reporting window that resulted in >24 hours for some mean values. ‡The per-protocol set excludes VIASKIN© Peanut Patches that were removed for reasons not related to adhesion.
The daily recording of system applications and removals, over the entire DBPC period, is referred to as the 'Global diary'.
ADWT, average daily wear time; DBPC, double-blind, placebo-controlled.